B-501a

Passive Immunization with RSV F Vaccine-Induced Polyclonal Antibodies Protect Cotton Rats From RSV A Challenge

R. Raghunandan, G. Smith, L. Fries, B. Zhou, H. Lu, D. Flyer, G. M. Glenn, MD — Novavax, Inc., Rockville, MD

Novavax
(240) 268-2000
ir@novavax.com

Abstract

Background: A candidate RSV F-protein nanoparticle vaccine has been evaluated in 3 human trials, towards development of a maternal vaccine to passively protect infants against RSV disease. High levels of palivizumab-competing antibodies (PCA) were observed in vaccines recipients[1]. The cotton rat (CR) challenge model provided supportive data for the successful palivizumab infant efficacy studies. We evaluated the ability of RSV F vaccine-induced polyclonal antibodies containing PCA to passively protect against RSV challenge.

Methods: Pooled sera from CRs immunized with RSV F vaccine were determined to have an RSV A neutralizing titer (MN) of 640, anti-F IgG titer of 2.4 x 10^6 and PCA equivalent of 844 µg/mL[2]. Female CR 4-6 weeks of age were passively immunized with 0.4, 0.1 or 0.04 mL of vaccine-induced immune serum, 0.4 mL of pre-immune serum, or 5, 1.25, or 0.625 mg/kg of palivizumab, given IM. One day later, animals were challenged intranasally with 10^5 pfu of RSV A, Long strain. Lungs were harvested on day 4 and homogenized for virus plaque assay.

Results: RSV F positive immune sera given IM at an 0.4 mL dose protected CR challenged one day later against detectable lung RSV replication. Palivizumab administered IM at 5 mg/kg was also protective, with lesser effects at lower doses. PCA and neutralizing antibodies were detected in passively-treated CR sera pre-challenge, and 50 µg/mL PCA and MN of 30 was associated with a >2 log viral load reduction.

Conclusions: Passive immunization with RSV F nanoparticle vaccine-induced polyclonal antibodies protected CR from RSV challenge. PCA levels that were associated with ~2 log reduction in viral load were similar to historical data on protective levels of palivizumab and suggest the antibody effect on viral lung titers, as measured by the PCA assay is comparable to historical measures of 'protective' palivizumab (25-30 µg/mL). The data demonstrate that the RSV F nanoparticle vaccine induces antibodies that given passively prevent RSV replication and at levels of palivizumab-like antibodies predictive of protection of infants in a maternal immunization regimen.

Table 1: Palivizumab-competing and neutralizing antibody levels and lung viral titers from cotton rats passively treated with vaccine-induced antibodies or Palivizumab

Antibody Groups	Lung Viral Titer (Log_{10})	RSV-Neutralization Titer (Log_2)	Palivizumab-Competing Antibody (µg)
Anti-RSV F Serum (0.4 mL)	<2.3[1]	6.1	179
Anti-RSV F Serum (0.1 mL)	2.5	4.9	51
Anti-RSV F Serum (0.04 mL)	2.9	3.7	<20[3]
Palivizumab (5 mg/kg)	<2.3[1]	5.5	68
Palivizumab (1.25 mg/kg)	3.7	3.5	20[3]
Palivizumab (0.625 mg/kg)	3.9	<3.3[2]	20[3]
Placebo	4.5	<3.3[2]	20[3]

Introduction

RSV continues to be a leading cause of respiratory illness related hospitalization in infants, children and the elderly. Passive prophylaxis using anti-F polyclonal and monoclonal antibodies have been shown to reduce disease burden in infants. A vaccine that generates similar immune responses may be effective in controlling RSV disease.

In clinical studies, the insect cell derived RSV F nanoparticle vaccine candidate has been shown to induce robust anti-F, PCA and MN antibody responses[1]. Similar responses are seen using the RSV F nanoparticle vaccine in cotton rats, which induces complete protection in live viral challenge[2]. The vaccine appears to be particularly efficient in the induction of anti-F antibodies that compete with palivizumab, a site II (Asn 258-Val 278) binding monoclonal antibody, currently used in passive prophylaxis in infants.

The RSV F nanoparticle vaccine is being developed in the context of maternal immunization, where placental transfer of antibody may protect young infants against severe RSV disease. The development of palivizumab was guided by the cotton rat model and human PK studies. In these studies trough levels of palivizumab at 25-30 µg/mL were successful used as guidance for field efficacy testing[3].

The RSV F nanoparticle vaccine has been evaluated in clinical studies and the induction of palivizumab competing antibodies (PCA) suggests that the vaccine may be effective. Evaluation of anti-sera from immunized cotton rats and palivizumab given passively in the challenge model presents an opportunity to determine whether the competitive ELISA measure benchmarks levels associated with protection and thus may be useful as a marker for protection in future efficacy studies.

Study Objectives

- To establish if passively administered anti-F antibodies could reconstitute active immunity in naive cotton rats
- To establish if prophylactically administered anti-F antibodies could protect naive cotton rats against live viral challenge
- To establish a dose response curve for anti-F passive prophylaxis in comparison to palivizumab

Study Design

Table 2: Study Design

Group	Antibody	Antibody Dose (rat)	Target Dose (palivizumab)
A	Anti-RSV F	0.4 mL	—
B	Anti-RSV F	0.1 mL	—
C	Anti-RSV F	0.04 mL	—
D	Palivizumab	—	5 mg/kg
E	Palivizumab	—	1.25 mg/kg
F	Palivizumab	—	0.625 mg/kg
G	Normal Cotton Rat Sera	0.15 mL	Negative Control

Figure 1: Study Design

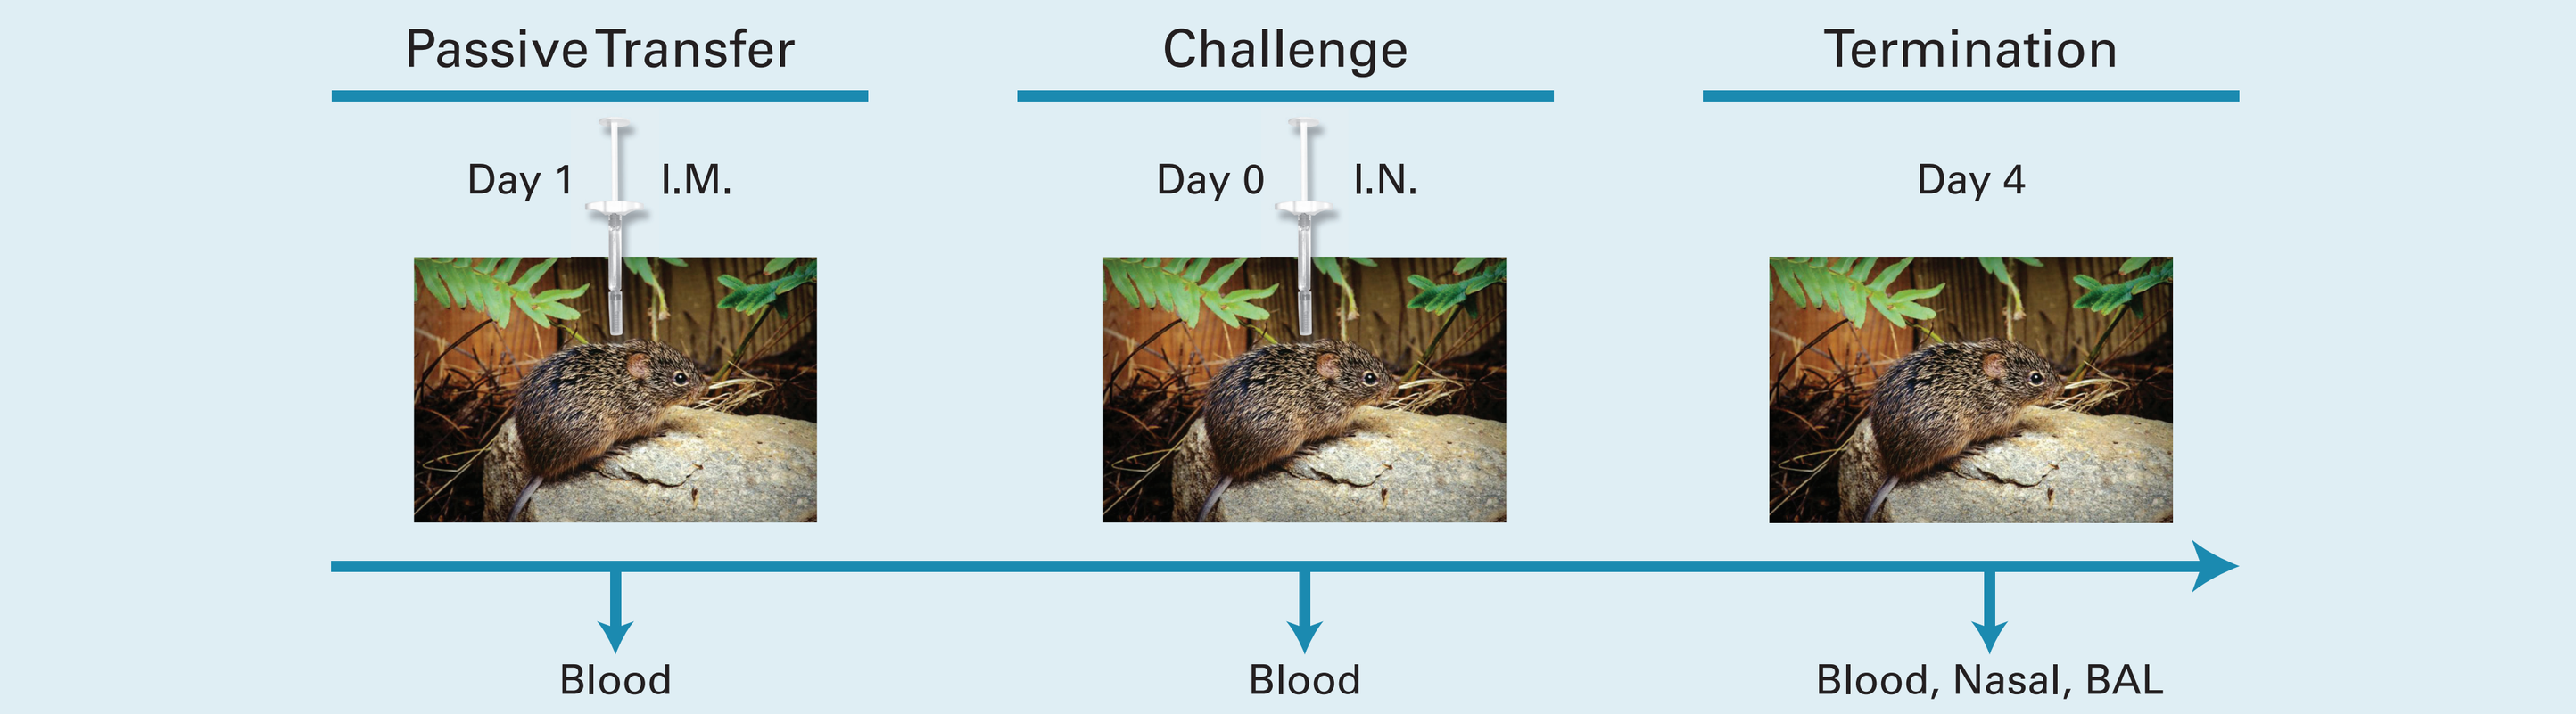
Passive Transfer
Challenge
Termination
Day 1
I.M.
Day 0
I.N.
Day 4
Blood
Blood
Blood, Nasal, BAL

- Female Cotton rats 4-6 weeks old were passively immunized by intra muscular injection with either anti-F immune sera (0.4, 0.1,0.04 mL) or palivizumab (5, 1.25 0.625 mg/kg) on Day 1
- Twenty four hours post passive immunization all rats were subjected to intranasal challenge with RSV A/Long virus (10^5 pfu/100 µL/rat)
- On day 4 all rats were exsanguinated after a terminal bleed. Lung and BAL were isolated for evaluation of antibody and virus titers

Methods

- In vivo animal procedures were carried out at Sigmovir Inc.
- Viral titers from homogenized lung tissue were determined by plaque assay performed at Sigmovir
- Following antibody analysis were carried out at Novavax[2]
 - Anti-F IgG ELISA
 - Palivizumab competitive ELISA
 - RSV A/Long micro neutralization assay

Figure 2: Anti-F Antibodies Post Passive Immunization in Sera and Lung Washes

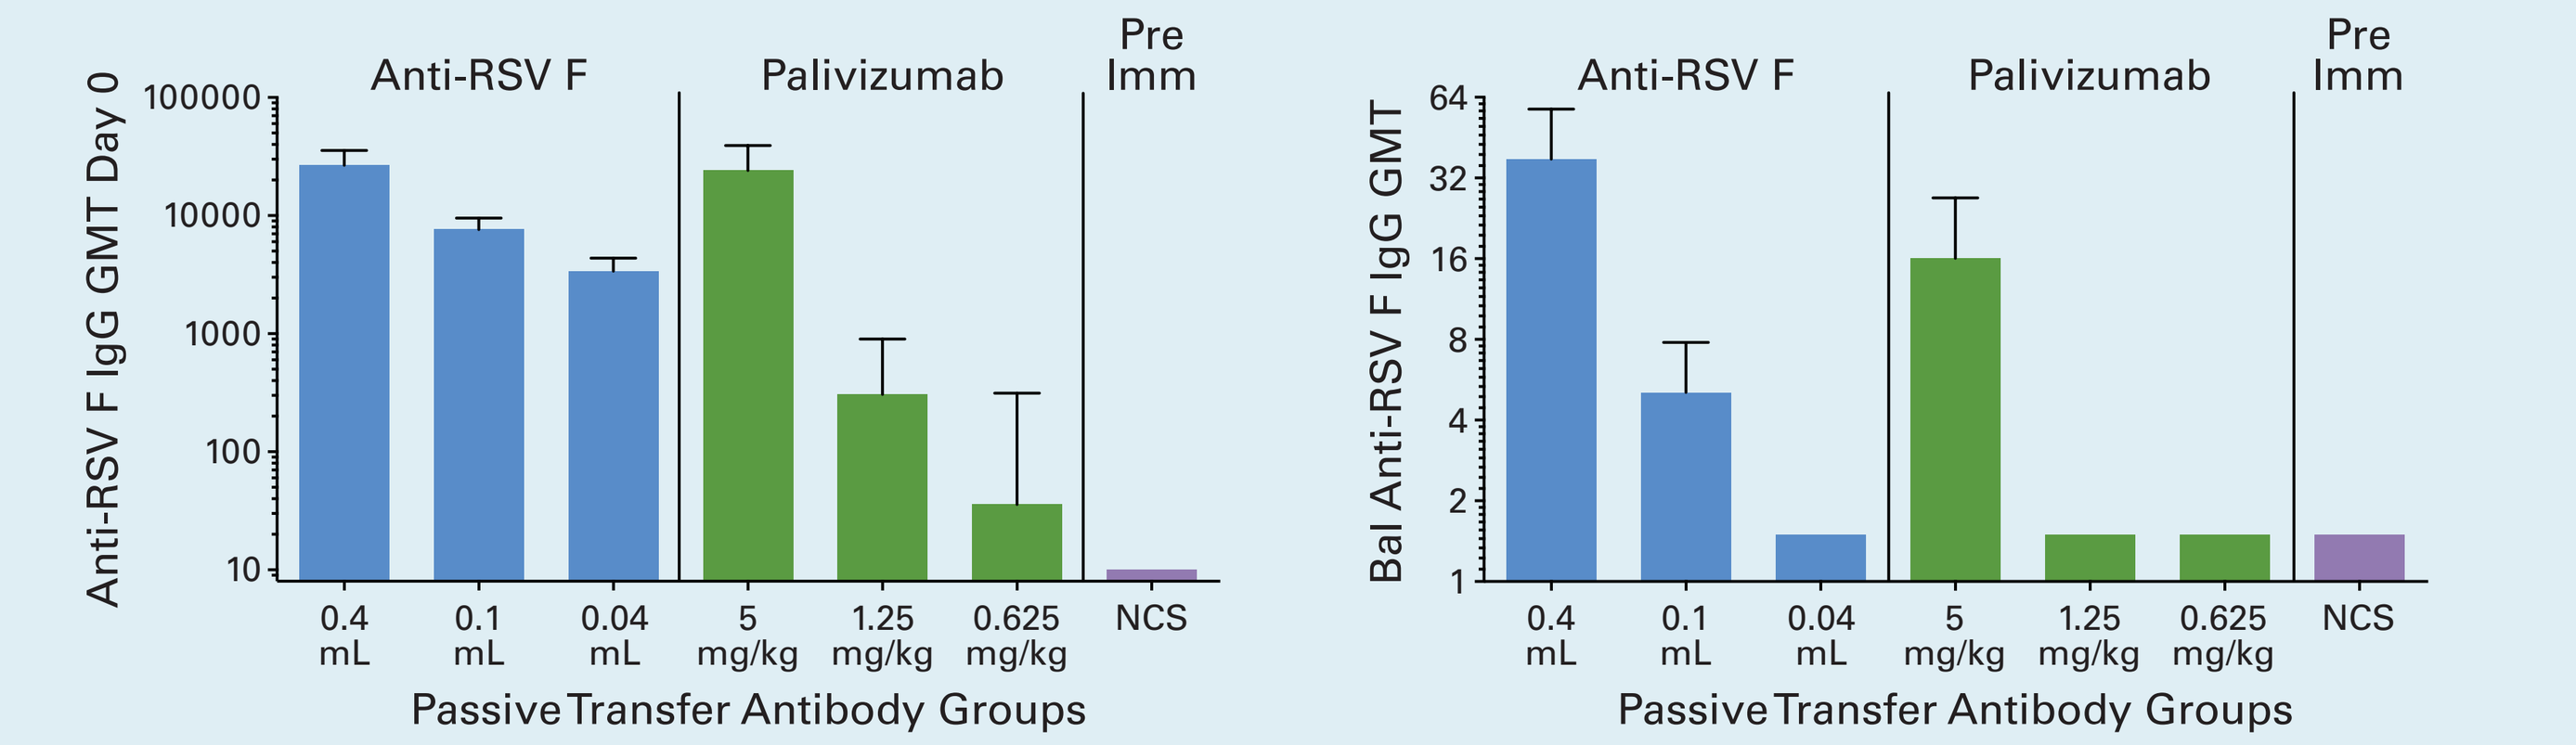
Anti-RSV F IgG GMT Day 0
Anti-RSV F
Palivizumab
Pre Imm
100000
10000
1000
100
10
0.4 mL
0.1 mL
0.04 mL
5 mg/kg
1.25 mg/kg
0.625 mg/kg
NCS
Passive Transfer Antibody Groups
Bal Anti-RSV F IgG GMT
64
32
16
8
4
2
1

Anti-RSV F IgG was evaluated by ELISA. Panel A: Day 0 serum. Panel B: BAL Geometric mean titers are plotted with 95% CI

Figure 3: Palivizumab Competing and MN Antibodies After Passive Immunization

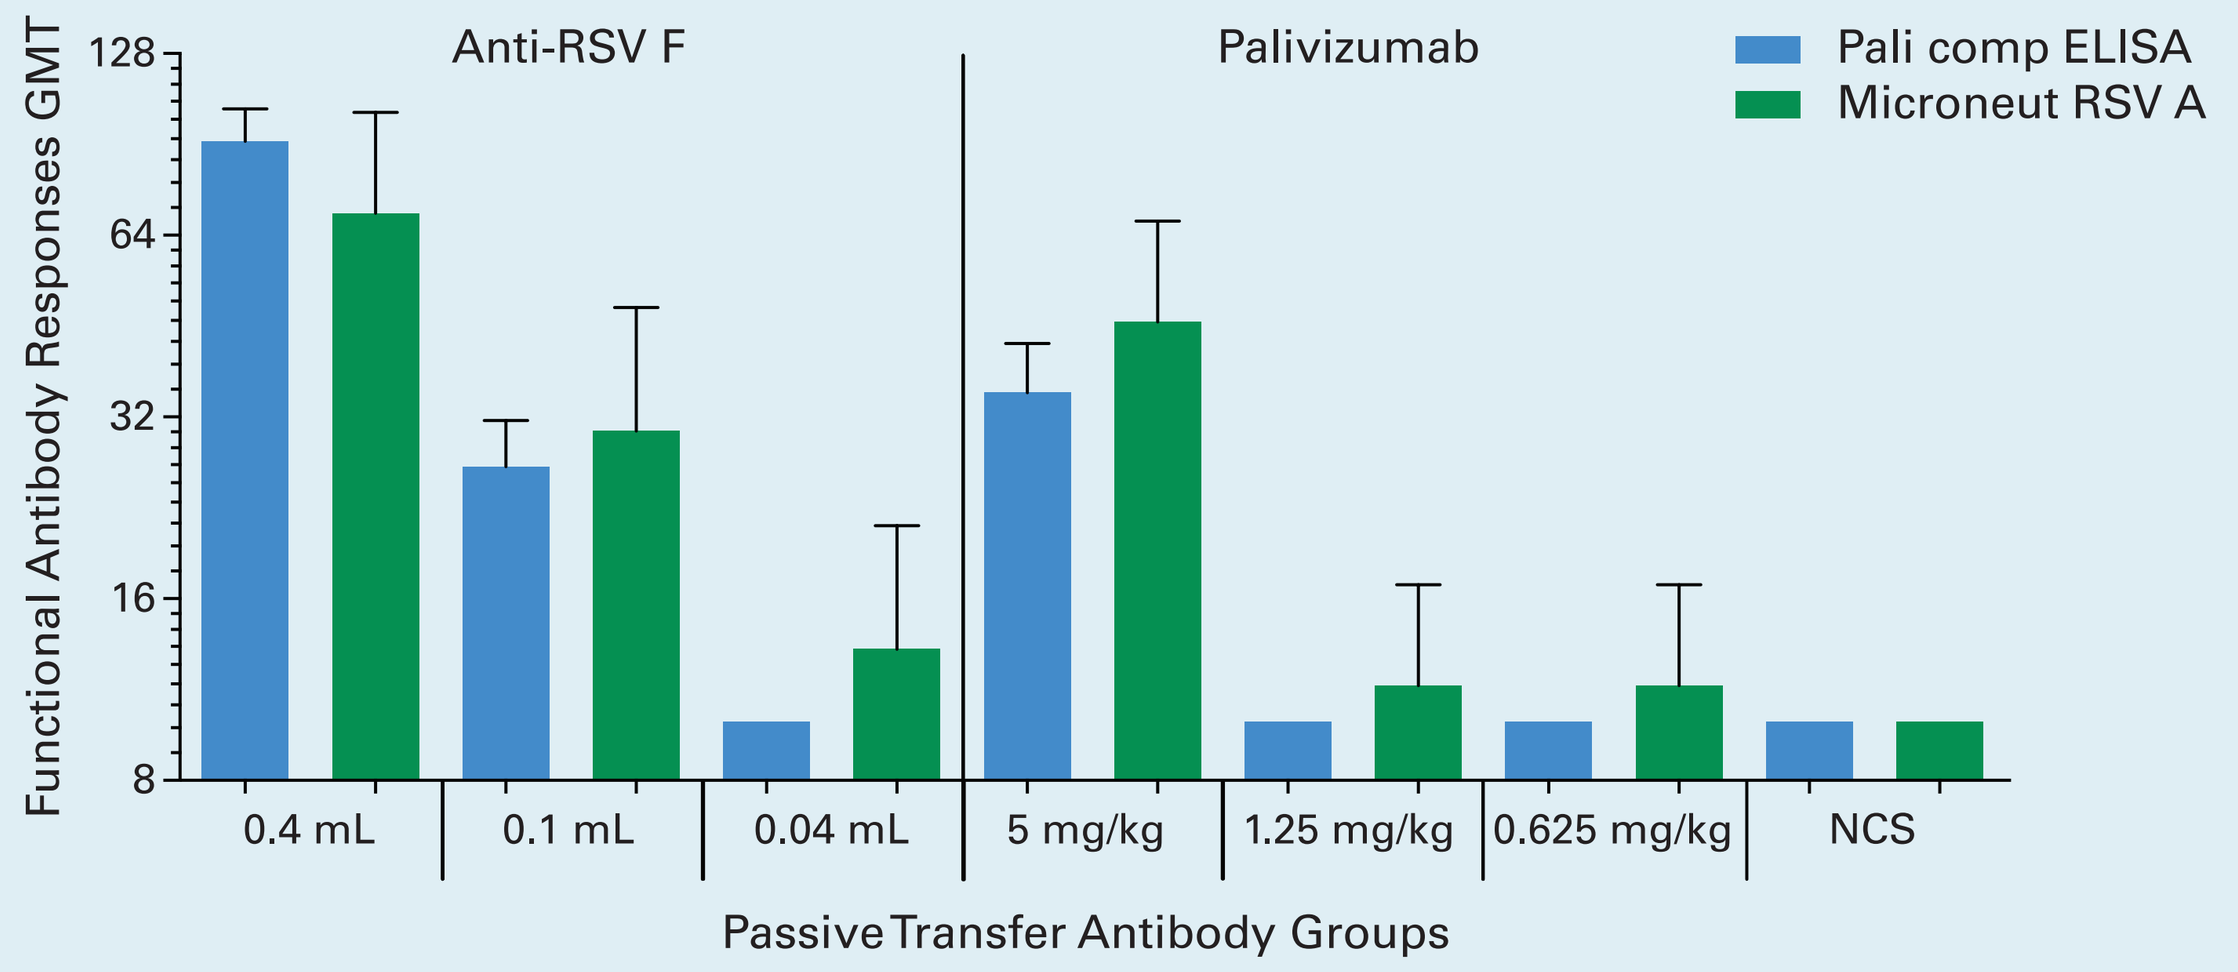
Functional Antibody Responses GMT
Anti-RSV F
Palivizumab
Pali comp ELISA
Microneut RSV A
128
64
32
16
8
0.4 mL
0.1 mL
0.04 mL
5 mg/kg
1.25 mg/kg
0.625 mg/kg
NCS
Passive Transfer Antibody Groups

Day 0 serum samples were subjected to palivizumab competitive ELISA and RSV A virus neutralization assay. Geometric mean titers at 50% inhibition of palivizumab competition and virus neutralization are plotted with 95% CI.

Immunogenicity Results

Passive immunization with vaccine-induced anti-F polyclonal antibody resulted in measurable anti-F IgG, PCA and MN antibodies that were similar to those observed after palivizumab administration.

Figure 4: Protection in Lung

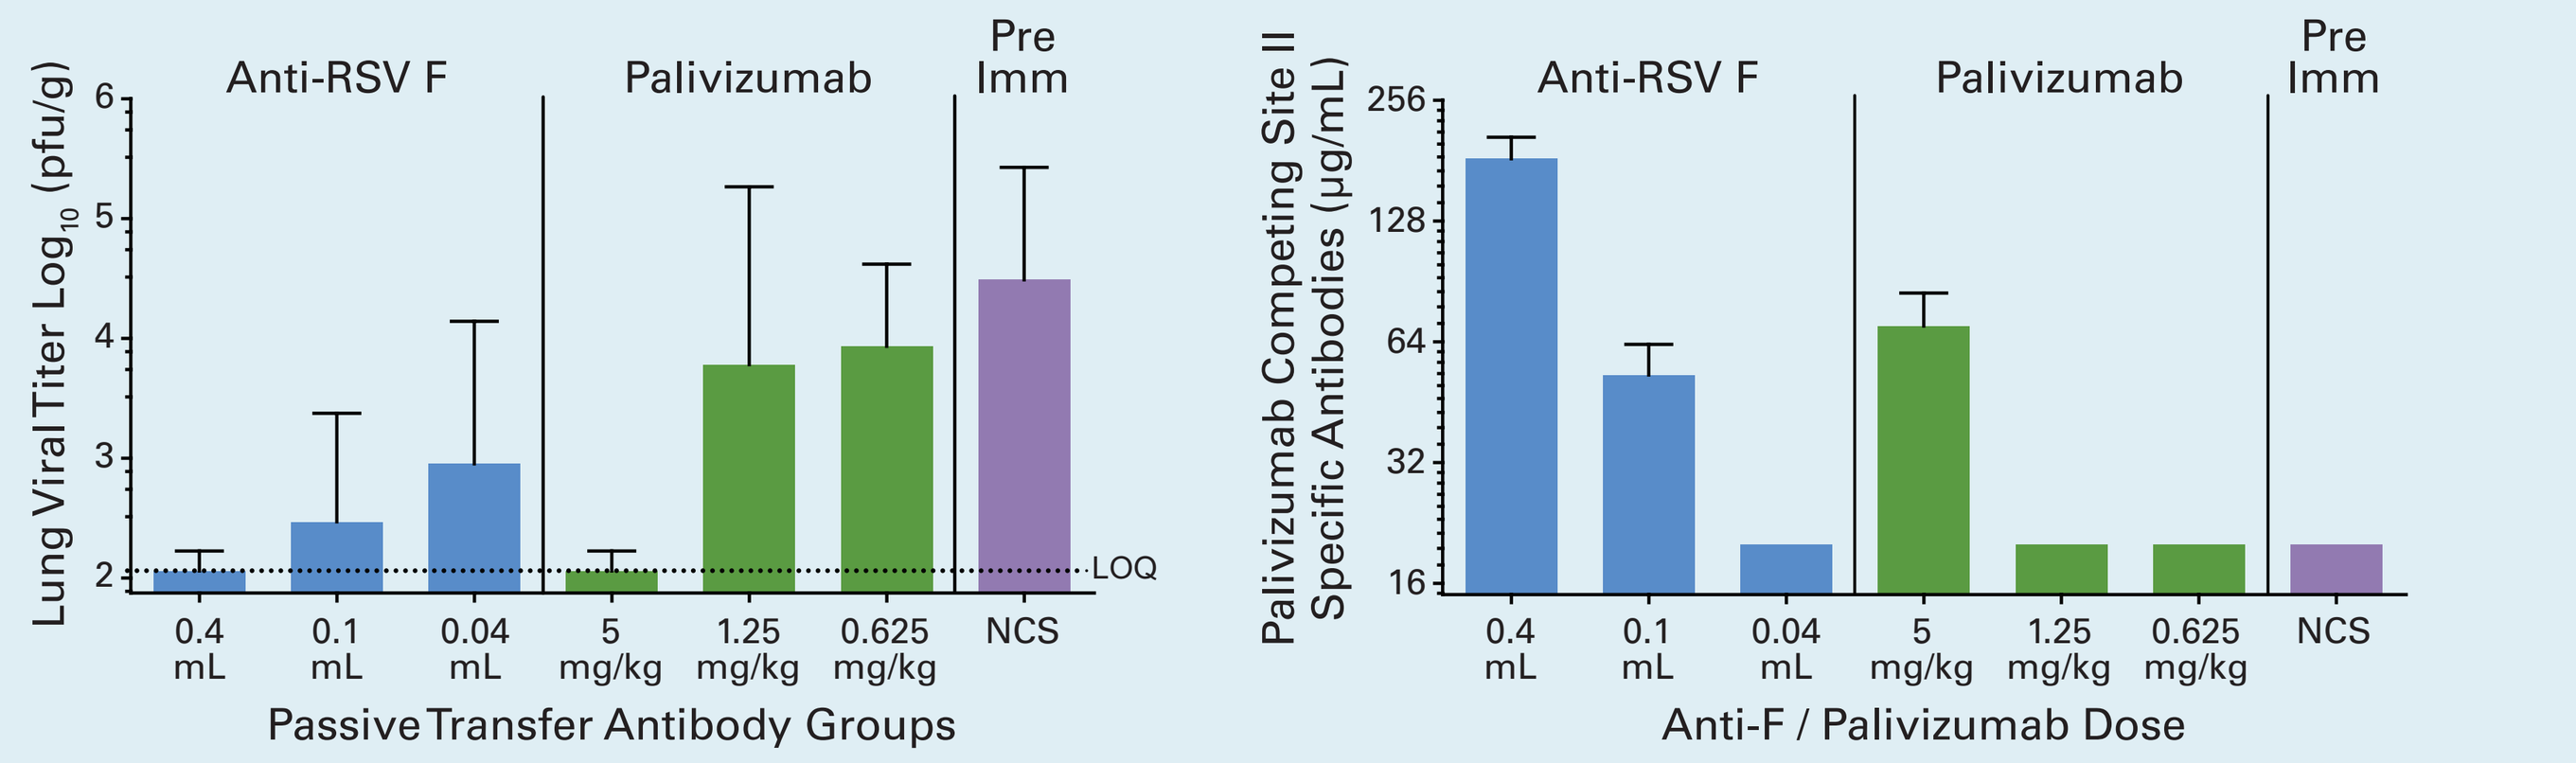
Lung Viral Titer Log10 (pfu/g)
Anti-RSV F
Palivizumab
Pre Imm
6
5
4
3
2
LOQ
0.4 mL
0.1 mL
0.04 mL
5 mg/kg
1.25 mg/kg
0.625 mg/kg
NCS
Passive Transfer Antibody Groups
Palivizumab Competing Site II Specific Antibodies (µg/mL)
256
128
64
32
16
Anti-F / Palivizumab Dose

Panel A: Day 4 post challenge viral lung tissue. Log_{10} titers were plotted with 95% CI.

Panel B: Palivizumab competing antibodies (µg/mL) in day 0 serum. 50% inhibition titer plotted with 95% CI.

Challenge Results

Passive immunization with polyclonal sera from animals immunized with the RSV F nanoparticle vaccine completely inhibited viral replication in the lung.

- Passive immunization with 100 µL of polyclonal sera resulted in a 2.0 log_{10} reduction in lung viral load and was associated with a palivizumab-like antibody of 51 µg/mL and MN titer of 4.9 log_2
- Passive immunization with 5mg/kg palivizumab completely inhibited viral replication in the lung and was associated with a palivizumab-like antibody of 68 µg/mL and MN titer of 5.5 log_2

Conclusions

- **Passive immunization with vaccine-induced anti-F polyclonal antibody protected cotton rats against live viral challenge**
- **Passive immunization with vaccine-induced anti-F polyclonal antibody resulted in measurable anti-F IgG, PCA and MN antibodies that were similar to those observed after palivizumab administration**
- **A PCA level of 51 mg/mL was associated with a 2.0 log_{10} reduction in lung viral load and was associated with a palivizumab-like and MN titer of 4.9 log_2**
- **This study suggests that PCA induced by the RSV F nanoparticle vaccine, and detected in the sera by the competition assay after passive administration, appears similar to the quantity of antibody associated in past studies to be protective in the cotton rat challenge model using palivizumab**

References: [1]Glenn et al, Vaccine, 31:524-32; [2]Smith G, et al. PLoS ONE 7: e50852; [3]Johnson et al, JID, 176:1215–24

Poster presented at the
53rd Interscience Conference on Antimicrobial Agents and Chemotherapy
September 10-13, 2013, Denver, Colorado